New therapies and clinical trials

LP025

Preclinical evidence for anti-inflammatory and immunomodulatory effects of NeuroRestore ACD856, a Trk-PAM in clinical development for the treatment of Alzheimer's disease.

Cristina Parrado-Fernández [1,2], Ruchi Gera [2], Veronica Lidell [1], Azita Rasti [1], Sumonto Mitra [2], Maria Backlund [1], Gunnar Nordvall [1,2], Martin Jönsson [1,2], Johan Sandin [1,2], Maria Eriksdotter [2,3], Pontus Forsell [1,2]

1. AlzeCure Pharma AB, Hälsovägen 7, Sweden
2. Division of Clinical Geriatrics, Department of Neurobiology, Care Sciences and Society, Karolinska Institutet, Stockholm, Sweden
3. Theme Inflammation and Aging, Karolinska University Hospital, Stockholm, Sweden

Objectives

The objective of these in vivo studies was to explore whether ACD856, a positive modulator of Trk-receptors, displays any immunomodulatory and anti-inflammatory effects in AD and other age-related disorders that would support potential disease-modifying effects of this novel molecule.

Background

Neurotrophins like BDNF, NGF, NT-3, and NT-4/5 are essential for neuron development and survival, acting through Trk receptors (TrkA, TrkB, TrkC). They play key roles in neurodegenerative diseases, including Alzheimer's Disease (AD), by supporting neuronal survival, plasticity, and cognition. NeuroRestore ACD856, developed by AlzeCure, is a positive allosteric modulator of Trk receptors, enhancing neurotrophin signaling. It has successfully completed phase I clinical trials, demonstrating very good safety and pharmacokinetics, and signals of CNS activity of ACD856. In in vivo models, ACD856 has demonstrated pro-cognitive effects, increased BDNF levels and long-lasting antidepressant-like effects. In vitro, ACD856 displayed enhanced neurite outgrowth and neuroprotective effects i.e. against Aβ toxicity, indicating both cognitive enhancing/symptomatic and disease modifying properties. Given that NGF, BDNF, and their Trk receptors are not only expressed by neurons, but also immune cells, microglia, and astrocytes, and are involved in modulating immune function, i.e. enhancing B- and T-cell survival and regulating cytokine and antibody production, we aimed to explore ACD856's potential immunomodulatory and anti-inflammatory effects in AD and other age-related disorders.

Methods

ACD856 was tested for effects on inflammatory processes in normal aged as well as APP mice. Aged male C57BL/6J mice (21-months-old) received daily subcutaneous ACD856 (5 mg/kg, n=12) or vehicle (n=11) for 28 days, with vehicle-treated 4-months-old mice (n=7) as young controls. In the AD model, 24 female APP-knock in mice ($APP^{NL\text{-}G\text{-}F}$ mice) and 12 female wild-type C57BL/6JRJ 10-11 months of age were given ACD856 (3 mg/kg, p.o.) or vehicle twice daily for 30 days. The health status and body weight were recorded every other workday. Both the plasma and brain were subsequently used for analysis of IgG, IgM and inflammatory markers determined by ELISA or western blot.

Results

Treatment with ACD856 for 4 weeks significantly reduced the levels of the pro-inflammatory mediators IL-6 and IL-1β in brains of aged mice, which were significantly increased as compared to young animals. Both IgG and IgM were also increased in the brain of old mice. Notably, ACD856 lowered light and heavy chain IgG protein levels in aged mouse brains. Likewise, total IgG showed a considerable rise in brain and plasma of aged mice that was substantially reduced by ACD856, while IgM remained unchanged. In $APP^{NL\text{-}G\text{-}F}$ mice, ACD856 also normalized increased brain IL-6 and plasma IgG levels to wild-type levels after 30 days of administration of the molecule, with no effect on IgM. This indicates that ACD856 may serve as an anti-inflammatory and immunomodulatory agent for AD and age-related diseases.

Effect of ACD856 on the immune response of aged male C57BL/6J mice

ACD856 reduces levels of IL-6 and IL-1β in brain

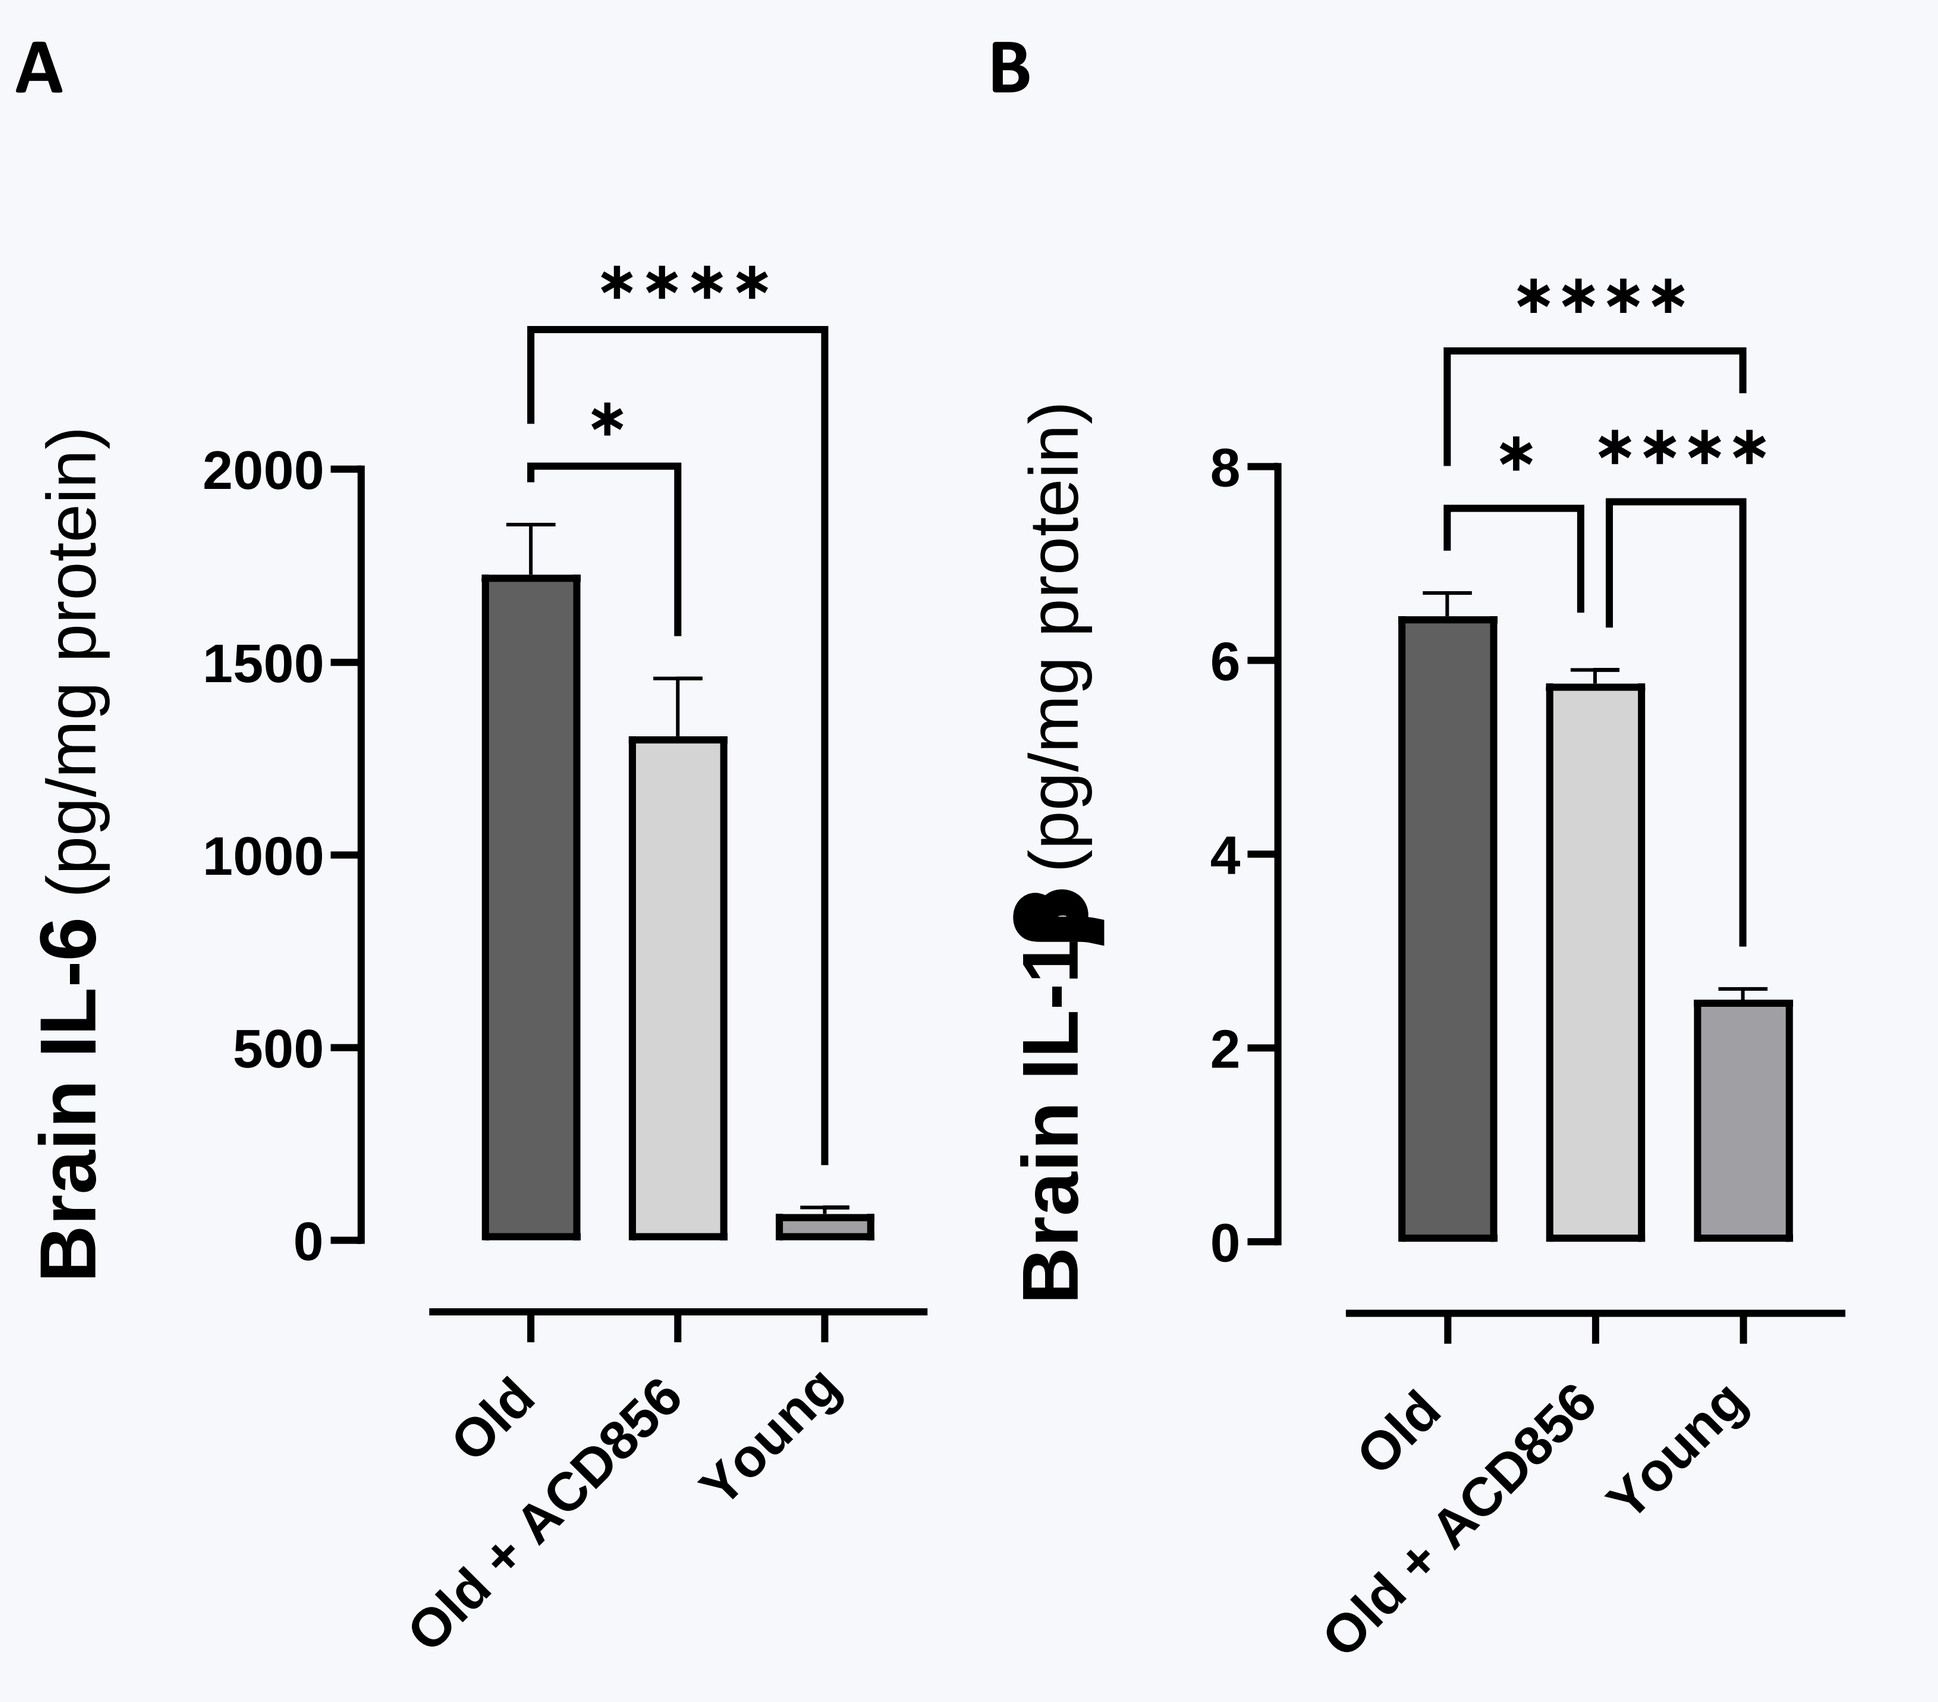
A
B
Brain IL-6 (pg/mg protein)
Brain IL-1β (pg/mg protein)
Old
Old + ACD856
Young

Figure 1. *Twenty-one months old mice were dosed with 5 mg/kg ACD856 once daily for 4 weeks by s.c. injection. The left hemisphere of each brain was homogenized, and the IL-6 (**A**) and IL-1β (**B**) levels were determined by ELISA. Data shown are the mean +/− SD, n = 11–12 animals. All samples were analyzed in duplicates in the ELISA assay, and cytokines levels were normalized to protein content. Significant differences between means were determined by one-way ANOVA, followed by Tukey's multiple comparisons test. * $p < 0.05$, **** $p < 0.0001$ vs vehicle-treated 4-months-old mice (young, n=7) and 21-months old mice (old) as control groups.*

ACD856 decreases levels of total IgG in both brain and plasma

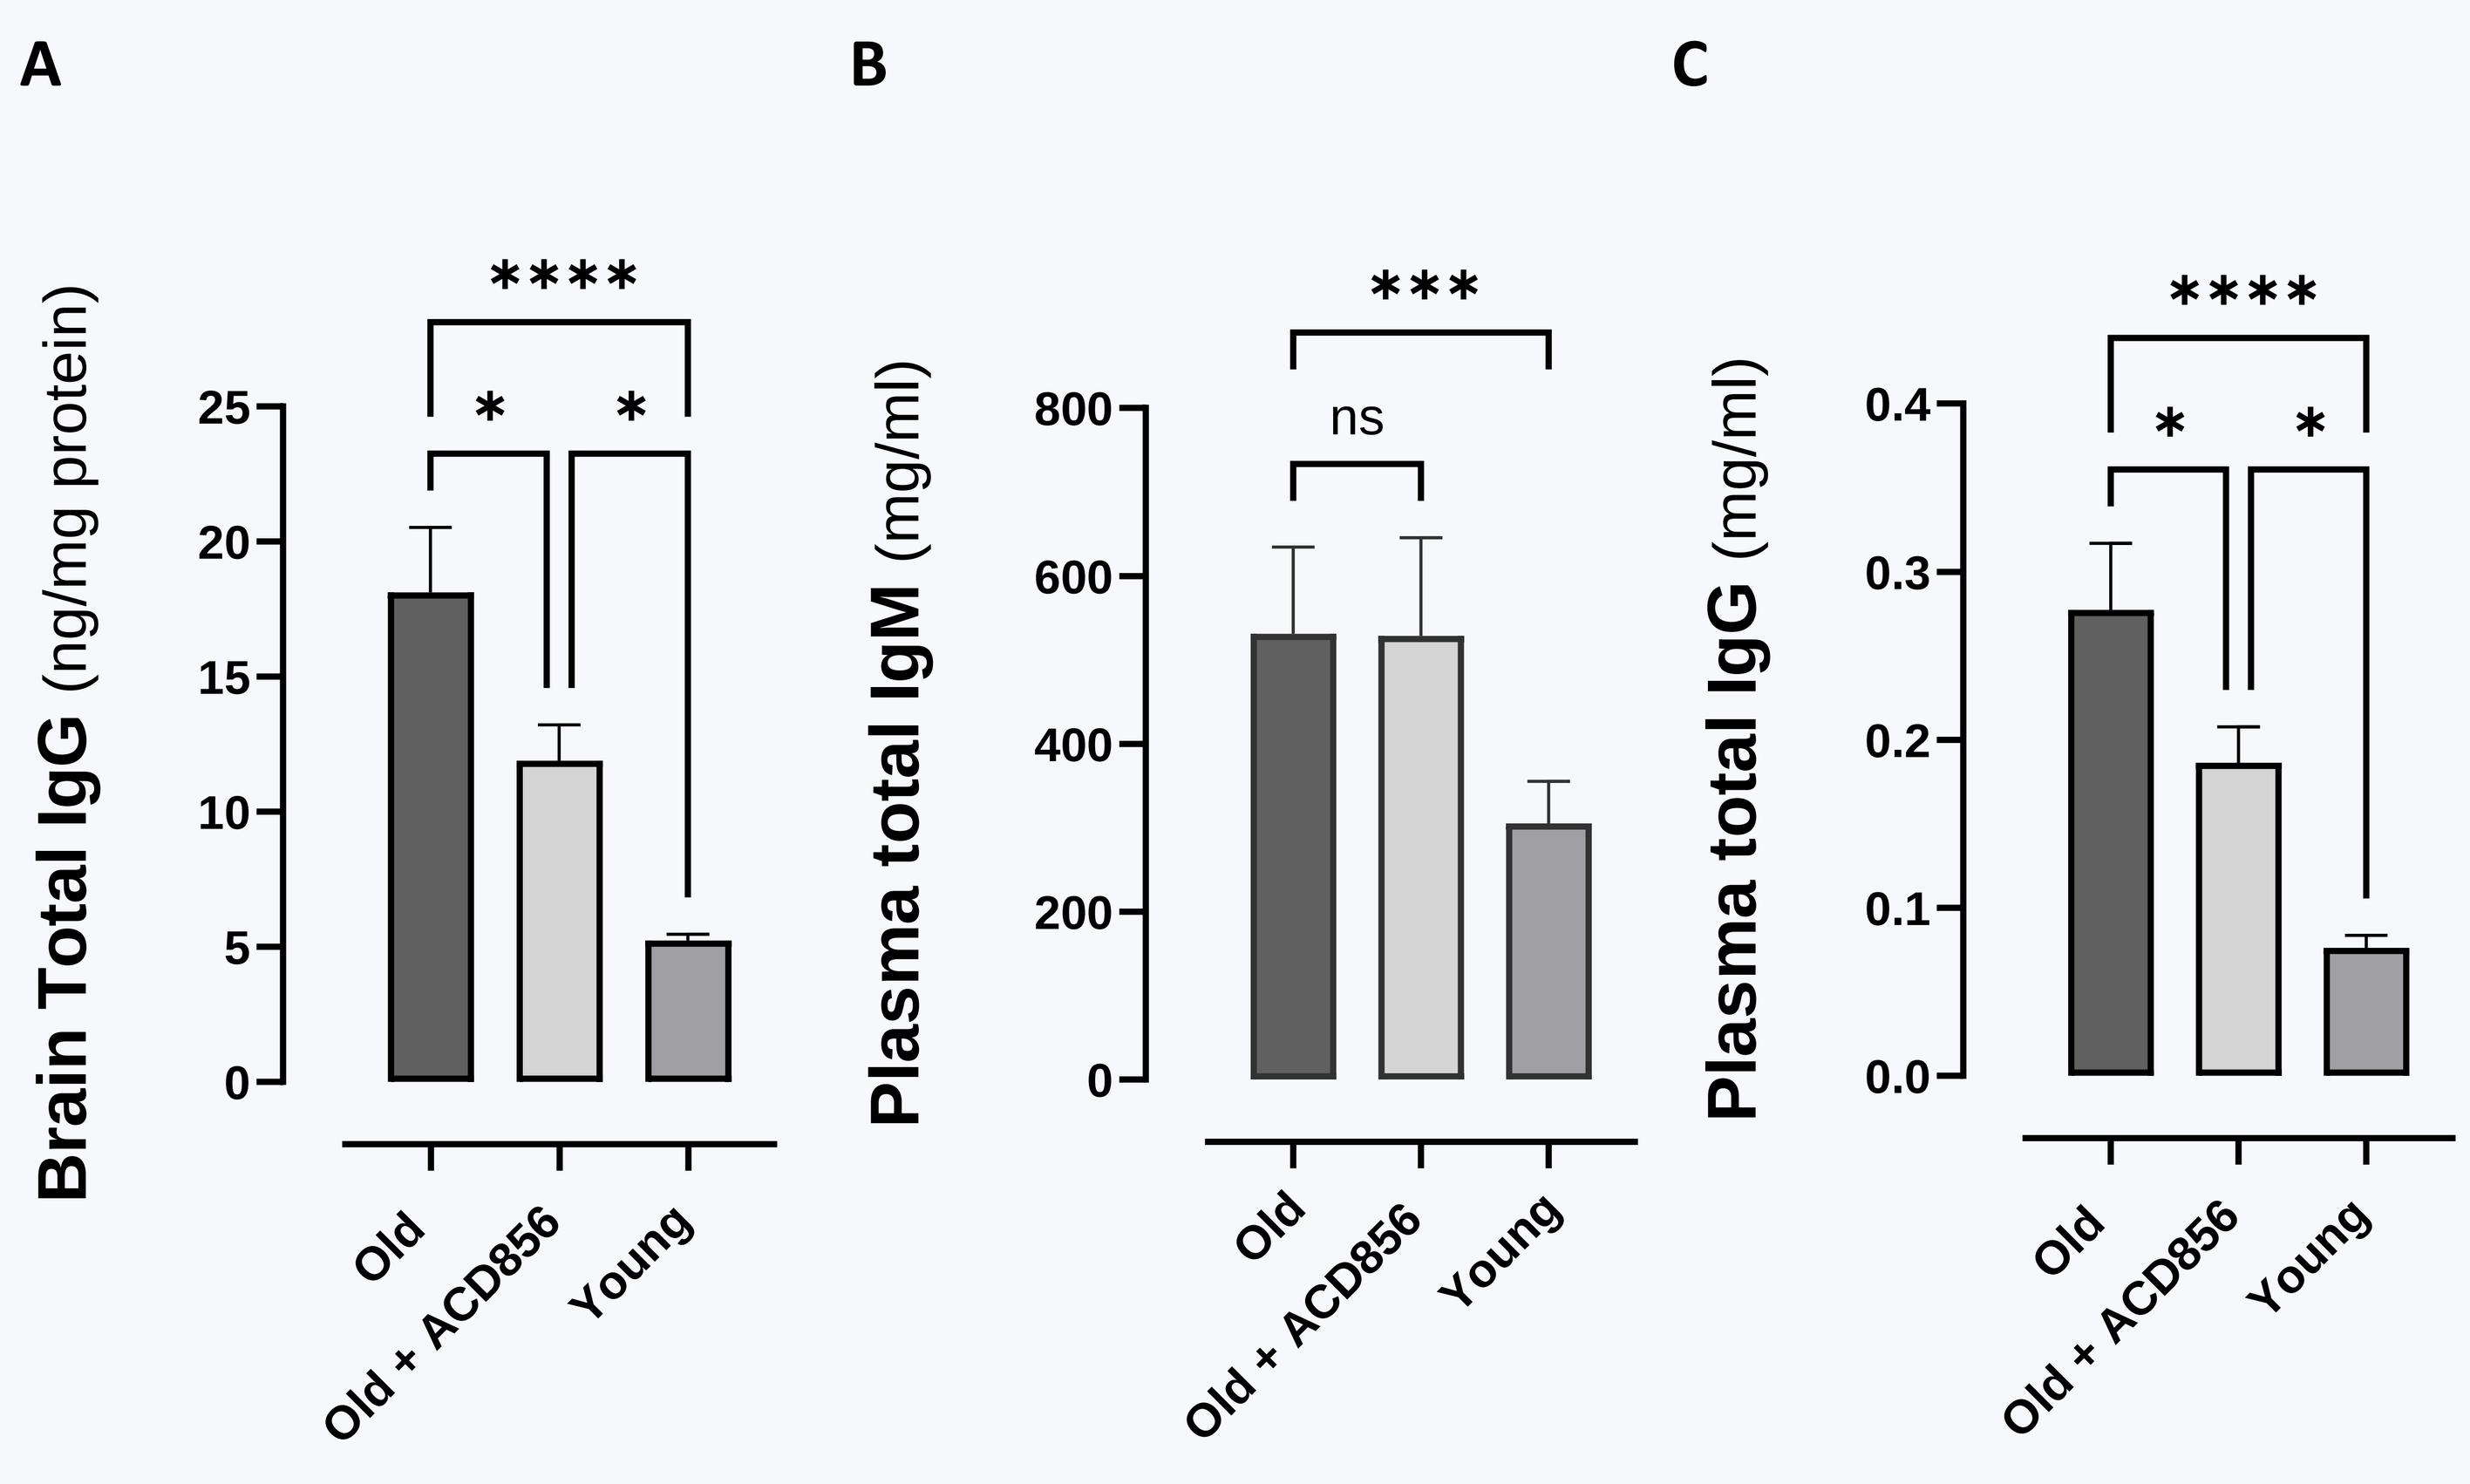
A
B
C
Brain Total IgG (ng/mg protein)
Plasma total IgM (mg/ml)
Plasma total IgG (mg/ml)
Old
Old + ACD856
Young

Figure 2. *Twenty-one months old mice were dosed with 5 mg/kg ACD856 once daily for 4 weeks by s.c. injection. Both the plasma and brain were subsequently used for analysis of IgG (**A,C**) and IgM (**B**) by ELISA. Data shown are the mean +/− SD, n = 11–12 animals. All samples were analyzed in duplicates in the ELISA assay. Significant differences between means were determined by one-way ANOVA, followed by Tukey's multiple comparisons test. * $p < 0.05$, *** $p < 0.001$ **** $p < 0.0001$ vs vehicle-treated 4-months-old mice (young, n=7) and 21-months old mice (old) as control groups.*

ACD856 lowers light and heavy chain IgG protein levels in aged mouse brains

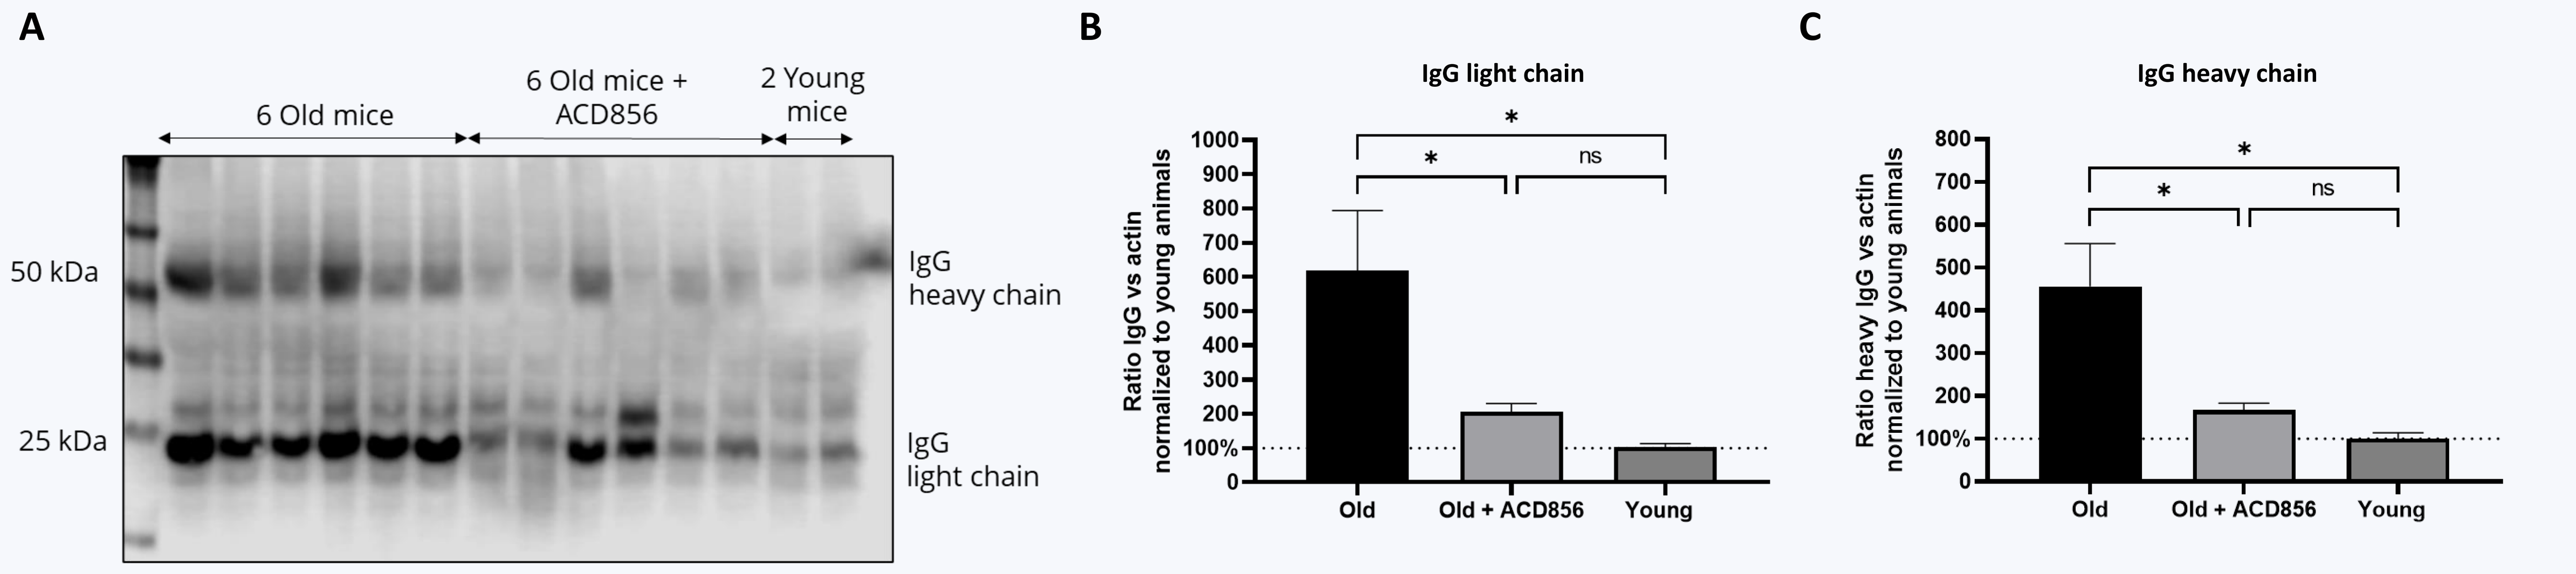
A
6 Old mice
6 Old mice + ACD856
2 Young mice
50 kDa
25 kDa
IgG heavy chain
IgG light chain
β-actin
B
IgG light chain
Ratio IgG vs actin normalized to young animals
C
IgG heavy chain
Ratio heavy IgG vs actin normalized to young animals
Old
Old + ACD856
Young

Figure 3. *Twenty-one months old mice were dosed with 5 mg/kg ACD856 once daily for 4 weeks by s.c. injection. The left hemisphere of each brain was homogenized and subsequently used for analysislysis of heavy and light chain IgG by western blot. Representative Western blot image of IgG heavy and light chains (**A**). Data shown for light chain (**B**) and heavy chain (**C**) of IgG are the mean +/− SEM of n=3 different gels, n = 11–12 animals. Significant differences between means were determined by one-way ANOVA, followed by Tukey's multiple comparisons test. * $p < 0.05$, vs vehicle-treated 4-months-old mice (young, n=7) and 21-months old mice (old) as controls.*

Effect of ACD856 on the immune response of APP^{NLGF} mice

ACD856 restores brain IL-6 to wild-type levels in APP^{NLGF} mice

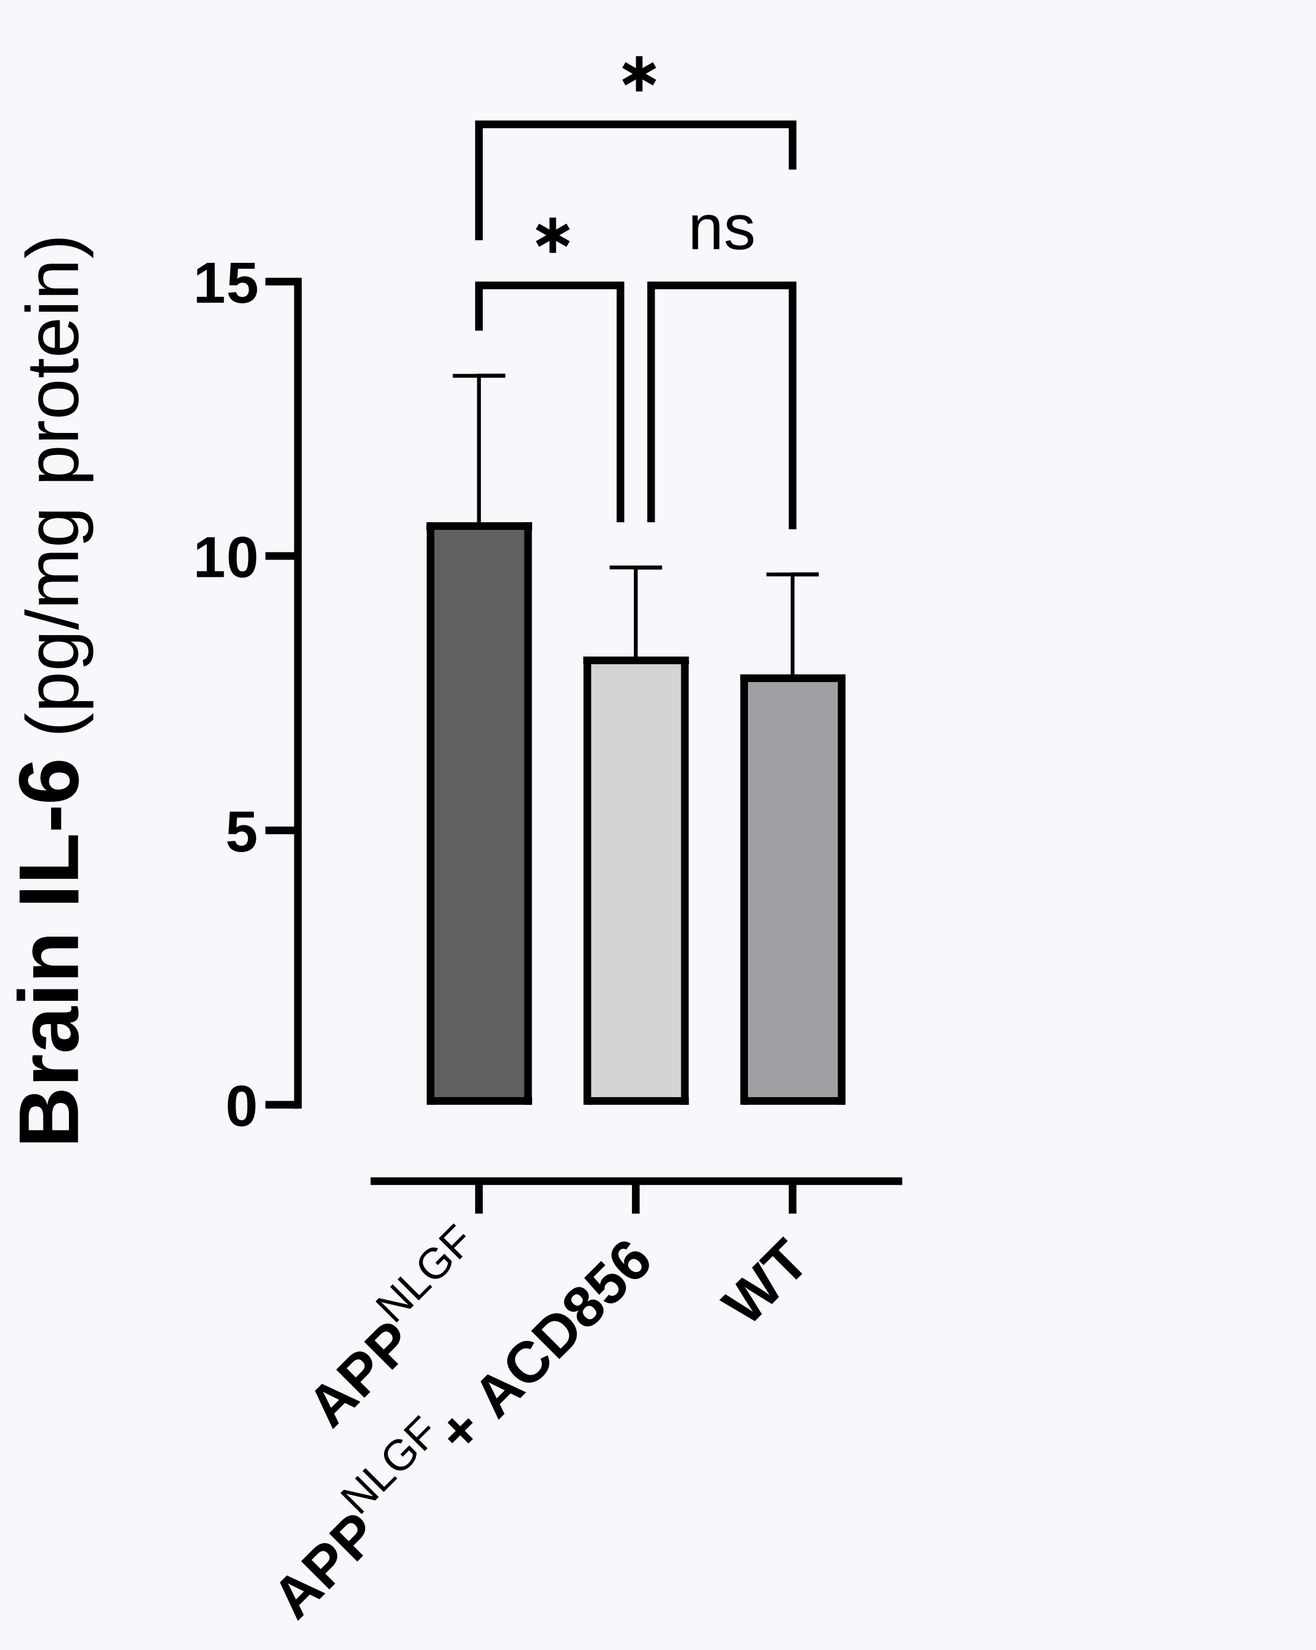
Brain IL-6 (pg/mg protein)
APP^NLGF
APP^NLGF + ACD856
WT

Figure 4. *Twenty-four female APP-knock in mice ($APP^{NL\text{-}G\text{-}F}$ mice) and 12 female wild-type C57BL/6JRJ 10-11 months of age were given ACD856 (3 mg/kg, p.o.) or vehicle twice daily for 30 days. The left hemisphere of each brain was homogenized, and the IL-6 levels were determined by ELISA. Data shown are the mean +/− SD, n = 11–12 animals. All samples were analyzed in duplicates in the ELISA assay, and IL-6 levels were normalized to protein content. Significant differences between means were determined by one-way ANOVA, followed by Tukey's multiple comparisons test. * $p < 0.05$, vs vehicle-treated mice (APP^{NLGF}) and wild-type (WT) as control groups.*

ACD856 restores plasma total IgG to wild-type levels in APP^{NLGF} mice

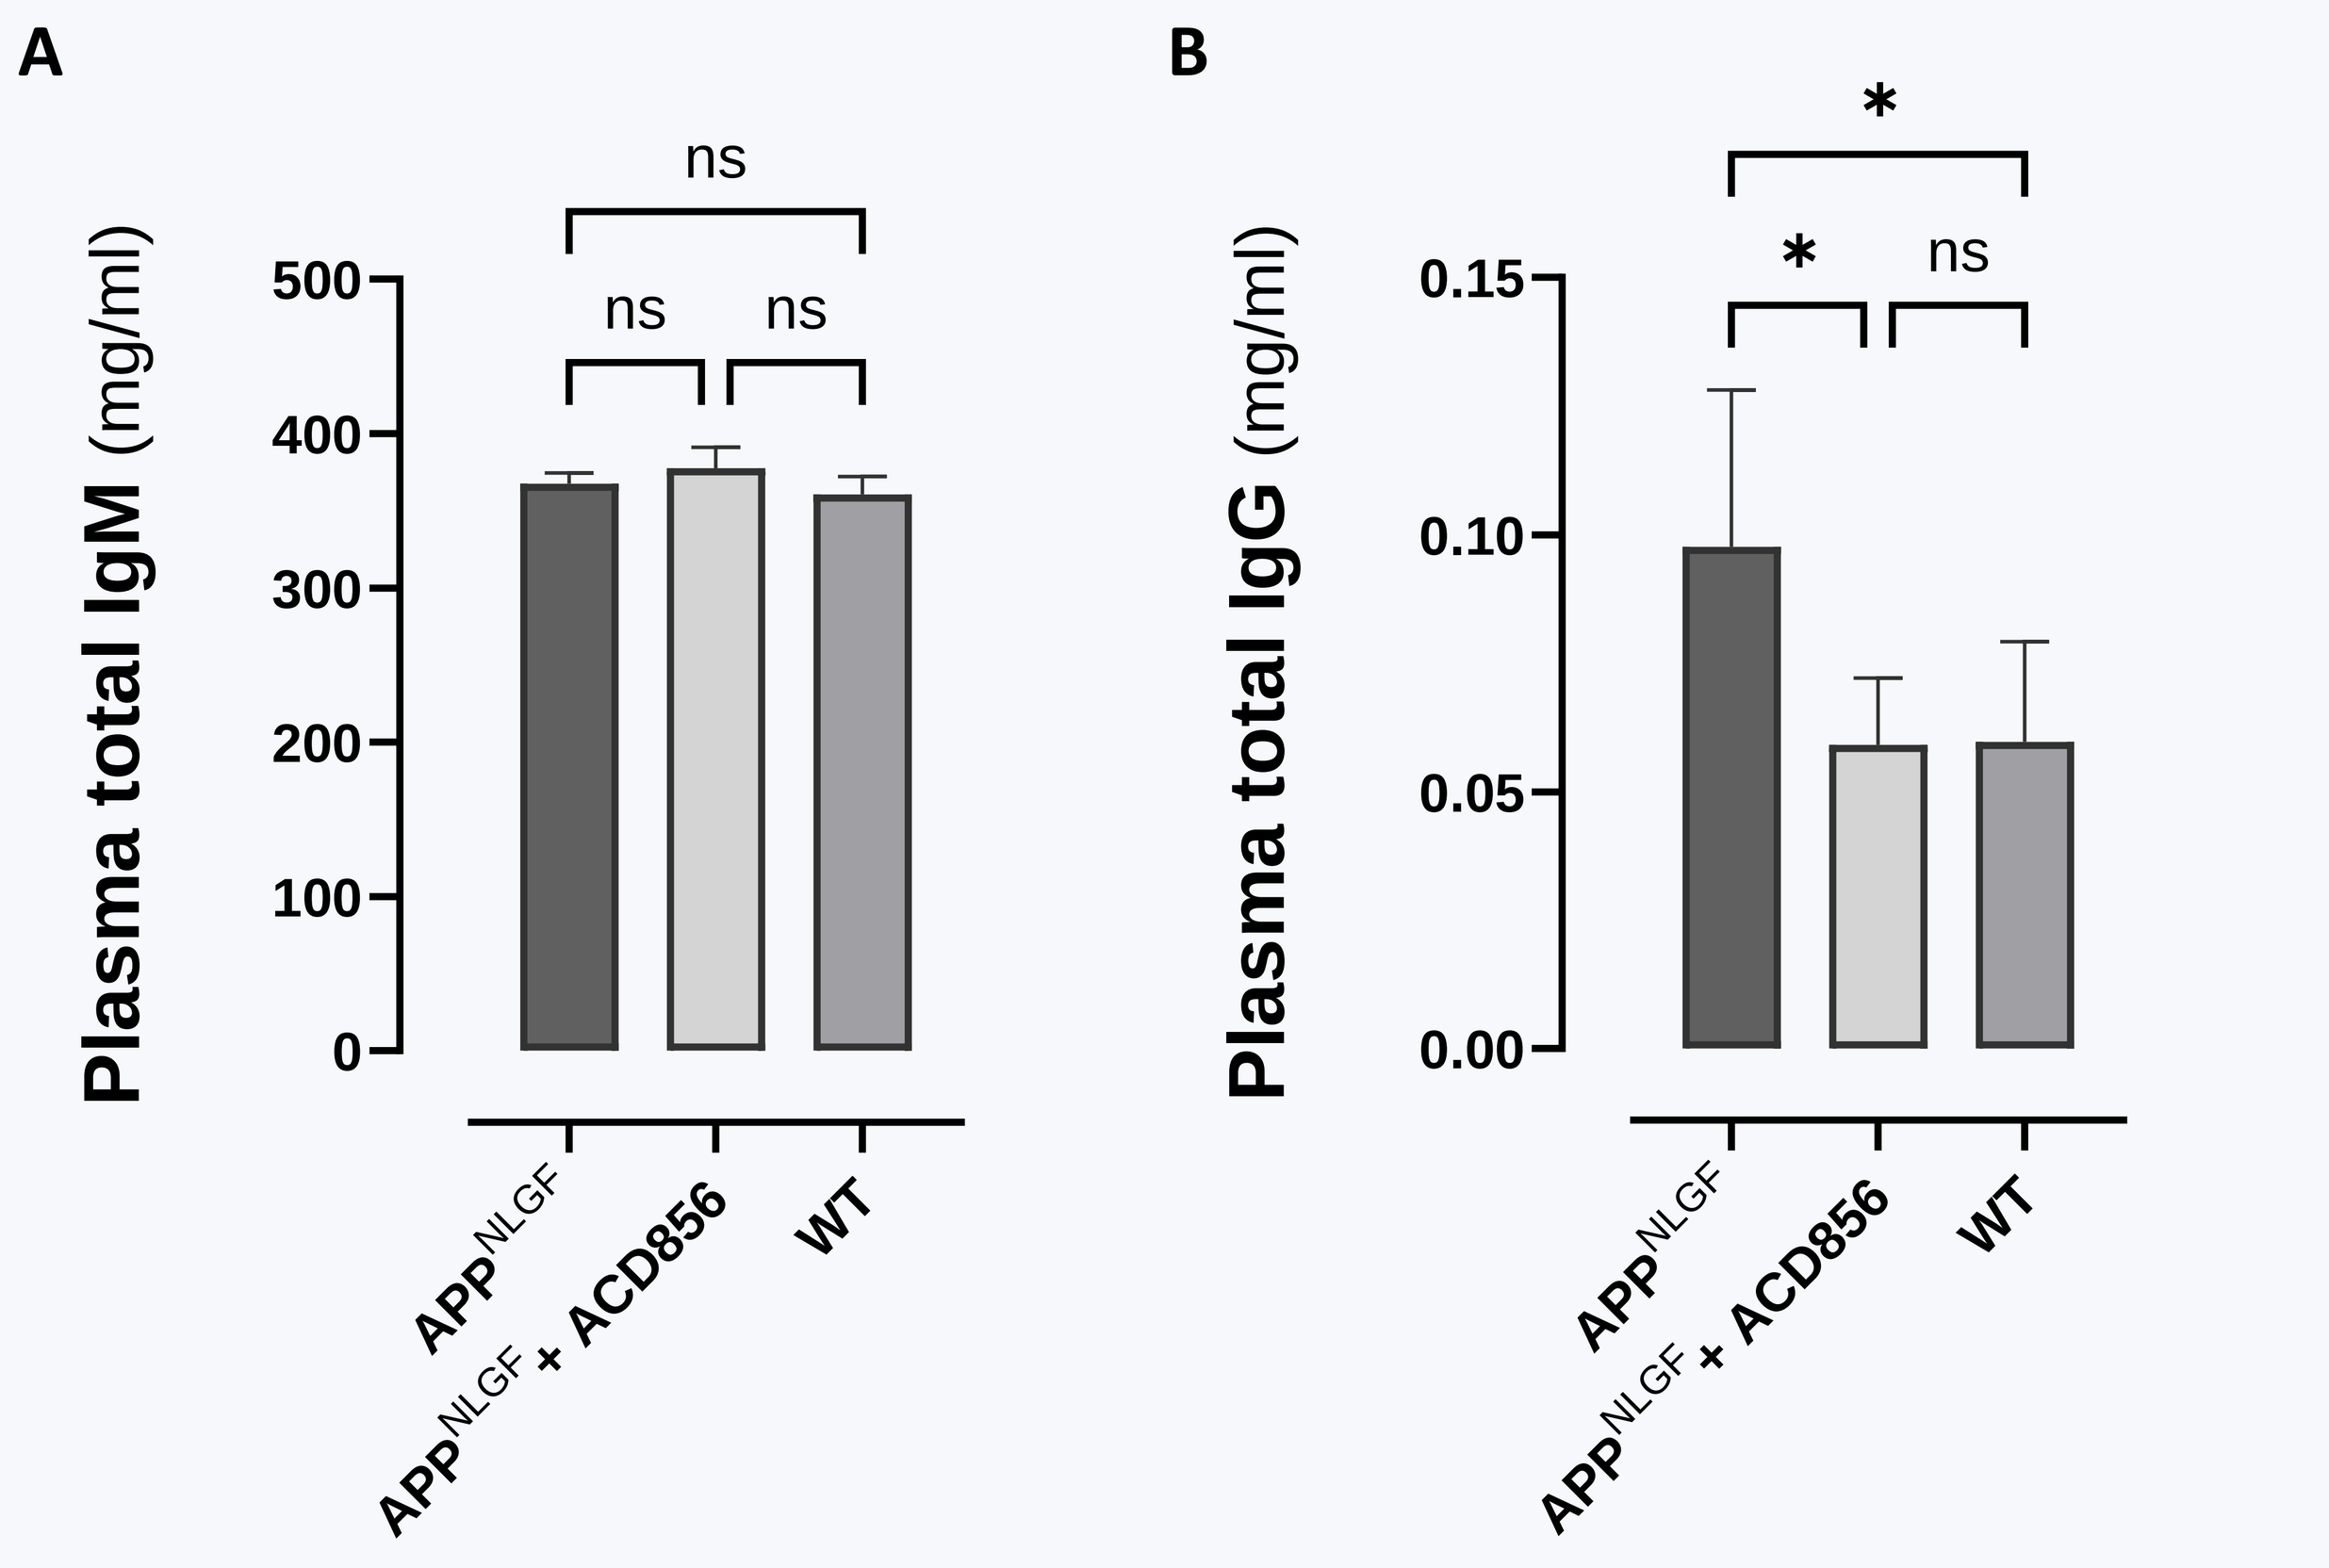
A
B
Plasma total IgM (mg/ml)
Plasma total IgG (mg/ml)
APP^NLGF
APP^NLGF + ACD856
WT

Figure 5. *Twenty-four female APP-knock in mice ($APP^{NL\text{-}G\text{-}F}$ mice) and 12 female wild-type C57BL/6JRJ 10-11 months of age were given ACD856 (3 mg/kg, p.o.) or vehicle twice daily for 30 days. Plasma total IgM (**A**) and IgG (**B**) levels were determined by ELISA. Data shown are the mean +/− SD, n = 11–12 animals. All samples were analyzed in duplicates in the ELISA assay. Significant differences between means were determined by one-way ANOVA, followed by Tukey's multiple comparisons test. * $p < 0.05$, vs vehicle-treated mice (APP^{NLGF}) and wild-type (WT) as control groups.*

Conclusion

The findings revealed a significant age and genotype effect on the immune response with higher levels of pro-inflammatory IL-6 and IL-1β cytokines and IgG and IgM when compared to respective control animals. Using NeuroRestore ACD856 to positively modulate Trk signalling we demonstrate a novel neuroprotective strategy by preventing altered secretion of inflammation-associated mediators. These findings suggest that ACD856 could play a novel anti-inflammatory and immunoregulatory role, alongside its cognitive-enhancing and disease-modifying effects, offering a promising strategy to slow neurodegeneration in AD and other diseases characterized by a neuroinflammatory component.

Copyright © 2024 Alzecure Pharma AB
Hälsovägen 7, 141 57, Huddinge, Sweden
martin.jonsson@alzecurepharma.com
www.alzecurepharma.com